Phase 1 Study of CTX-471, a Novel CD137 Agonist Antibody, in Patients with Progressive Disease Following PD-1/PD-L1 Inhibitors in Metastatic or Locally Advanced Malignancies: Monotherapy Dose Escalation and Dose Expansion Results.

#2535

AUTHORS: M. Barve[1], T. Johanns[2], N.Mettu[3], S. Rosen[4], J. Edenfield[5], M. Gutierrez[6], M. Giorlando[7], T. Schuetz[8]

Affiliations: [1]Mary Crowley Cancer Research, [2]Washington University Medical Center, [3]Duke University Medical Center, [4]Hematology Oncology Associates of the Treasure Coast, [5]Prisma Health Cancer Institute, [6]Hackensack University Medical Center, [7]Medical & Scientific Services Hematology/Oncology, IQVIA RDS Argentina, [8]Compass Therapeutics.

Abstract

Background: CTX-471, a fully human immunoglobulin G4 (IgG4) anti-CD137 agonist antibody, binds to a distinct epitope on CD137, and binds to the target with intermediate affinity which results in optimal agonism of the receptor and improved activation of T-cells and natural killer cells. Extensive preclinical studies have demonstrated potent antitumor activity of CTX-471, used as monotherapy or in combination therapy. CTX-471-001 (NCT03881488) is an ongoing phase 1 study that evaluates the safety and tolerability of CTX-471 alone and in combination with pembrolizumab. This report presents safety and efficacy data from the monotherapy arm, covering dose escalation and expansion cohorts.

Methods: This Phase 1, open-label, first-in-human study evaluates CTX-471 as monotherapy or in combination with pembrolizumab in patients with metastatic or locally advanced malignancies that have progressed while receiving an approved PD-1 or PD-L1 inhibitor. The monotherapy portion of the study has two parts: Part 1 Dose Escalation and Part 2 Dose Expansion. Monotherapy dose escalation ranges from 0.1-1.2 mg/kg IV biweekly, while Dose Expansion explores two dose levels: 0.3 and 0.6 mg/kg. The primary objective is to evaluate the safety and tolerability of CTX-471, with secondary objectives including PK, immunogenicity, and clinical activity.

Results: As of January 19, 2024, 19 patients were treated in the dose escalation and 60 patients were treated in the expansion part. 63% were male, and the median age was 66 years. Most common tumor types included non-small cell lung cancer (NSCLC) (25%), head and neck squamous-cell carcinoma (HNSCC) (21%), and melanoma (15%). In the 60 patient Dose Expansion cohort, there were patients with 17 different malignancies. The dose limiting toxicity observed in the dose escalation portion at 1.2 mg/kg was grade 4 thrombocytopenia. Treatment Related Adverse Events (TRAE) were reported in 64% of patients (51/79 of patients), and 87% of them were Grade 1-2. Treatment discontinuation due to AE was reported in 5 patients. Notably, a Complete Response (CR) was confirmed by PET scan in 1 of 3 patients treated with small-cell lung cancer. This patient, treated in the third-line setting, had a durable Partial Response (PR) for approximately 3 years prior to converting to a CR. Four additional PRs were also observed: 3 of 11 (27.3%) patients with melanoma and 1 of 4 (25%) patients with mesothelioma.

Conclusions: In this phase 1 study, CTX-471 was shown to be a safe and well-tolerated, novel anti-CD137 antibody. CTX-471 monotherapy demonstrates promising monotherapy anti-tumor activity in refractory patients who have progressed on approved PD-1 or PD-L1 inhibitors. A combination study with pembrolizumab is ongoing.

Introduction

- Immune checkpoint inhibition with antibodies targeting programmed cell death 1 (PD-1) and programmed cell death ligand 1 (PD-L1) have been transformational therapies, but a significant proportion of patients either do not respond to these agents or progress following an initial response or stable disease. Novel immunotherapies are clearly needed.
- However, the development of agonist antibodies targeting activating receptors has been challenging. CD137 (4-1BB) is an important costimulatory receptor on activated T and NK cells. Several drugs in development targeting CD137 have been limited by hepatic toxicity.
- In extensive preclinical studies, CTX-471 was discovered to be a unique agonist of CD137. The antibody binds to a unique epitope and intermediate affinity antibodies were optimal in preclinical efficacy studies.
- CTX-471-001 is a phase 1 study that evaluated the safety and tolerability of CTX-471 in patients who had previously received a PD-1 or PD-L1-containing regimen.
- This report presents safety and efficacy data from the covering dose escalation and expansion cohorts.

Clinical Trial information: NCT03881488, Research Sponsor: Compass Therapeutics, Inc.

Methods

Phase 1, open-label, first-in-human study of CTX-471: Dose Escalation and Dose Expansion

- Planned dose escalation ranged from 0.1 to 1.2 mg/kg IV biweekly, while Dose Expansion explored two dose levels: 0.3 and 0.6 mg/kg IV biweekly.
- Dose-limiting toxicities evaluated in a standard "3+3" design.

➢ Primary objective

- ➢ To evaluate the safety and tolerability of CTX-471.

➢ Key Eligibility Criteria

- ➢ Histologically confirmed diagnosis of metastatic or locally advanced malignancies.
- ➢ Disease progression after at least 12 weeks and at least 2 doses of a labeled PD-1 or PD-L1 inhibitor regimen per approved prescriber's information, whether as a monotherapy or in combination therapy, with no other intervening systemic anticancer therapy prior to enrollment in this study.
- ➢ ECOG 0 or 1
- ➢ Measurable disease per RECIST 1.1

Figure 1. Distribution of Treatment Related Adverse Events (TRAEs) by Grade

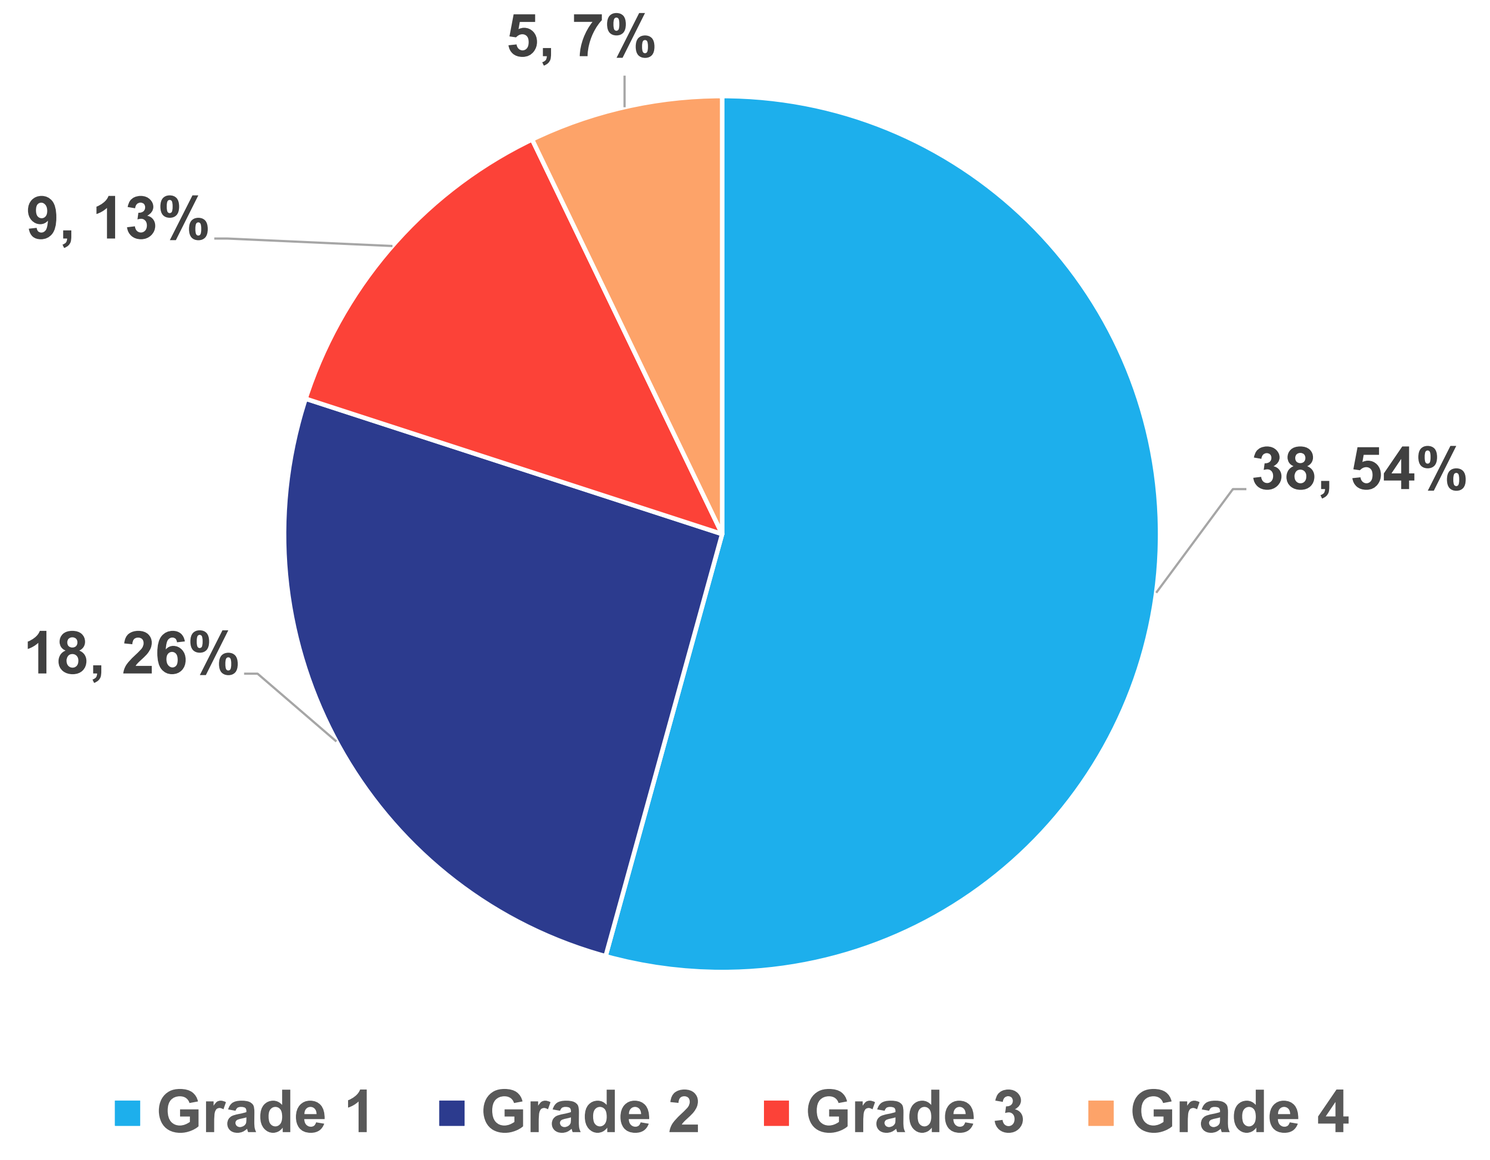
5, 7%
9, 13%
38, 54%
18, 26%
Grade 1
Grade 2
Grade 3
Grade 4

Results

Table 1. CTX-471-001 Baseline Characteristics

Attribute	Classes	Count (%)
Sex	Male	50 (63.2)
	Female	29 (36.8)
Tumor Type	NSCLC	20 (25.3)
	Melanoma	17 (21.5)
	HNSCC	13 (16.5)
	RCC	5 (6.3)
	Endometrial	4 (5.1)
	SCLC	3 (3.8)
	UC	2 (2.5)
	Merkel Cell Carcinoma	2 (2.5)
	Other	13 (16.5)
Age	≤65	36 (45.5)
	>65	43 (54.5)
ECOG	0	30 (37.9)
	1	49 (62.1)
Race	Caucasion	67 (84.8)
	Non-Caucasian	12 (15.2)

Table 1. As of May, 2024, 19 patients were treated in the dose escalation and 60 patients were treated in the dose expansion parts. 63% were male, and the median age was 66 years. Most common tumor types included non-small cell lung cancer (NSCLC) (25%), melanoma (21%) and head and neck squamous-cell carcinoma (HNSCC) (16%).

Table 2. TRAEs, SAEs, and DLTs

	Adverse Event	79 Patients N=# of Patients (%)
Most Common Treatment Related Adverse Events (All Grades)	Fatigue	18 (23)
	Nausea	6 (7.6)
	Pyrexia	6 (7.6)
	Anemia	5 (6.3)
	AST Increased	5 (6.3)
Treatment Related Serious Adverse Events	Pneumonitis/Hypoxia	1 (1.3)
	Thrombocytopenia	2 (2.5)
	Cytokine Release Syndrome	2 (2.5)
	AST/ALT Increased	1 (1.3)
Dose-Limiting Toxicity (1.2 mg/kg)	Thrombocytopenia	2 patients at 1.2 mg/kg

Figure 1 and Table 2. Treatment Related Adverse Events (TRAEs) were reported in 66% of patients (52/79 of patients), and 80% of them were Grade 1-2. The dose limiting toxicity observed in the dose escalation portion of the study occurred at 1.2 mg/kg and was grade 4 thrombocytopenia, seen in 2 patients. There were no grade 5 TRAEs in the study. The most common TRAEs for all grades were fatigue (18 patients, 23%), nausea (6 patients, 7.6%), pyrexia (6 patients, 7.6%), anemia (5 patients, 6.3 %), and AST increased (5 patients, 6.3 %). Among the 79 patients treated, there were 9 drug-related Serious Adverse Events (SAEs) that were observed in 4 patients: pneumonitis in one patient; thrombocytopenia and immune thrombocytopenic purpura in one patient; thrombocytopenia, ALT increased, AST increased, and cytokine release syndrome in one patient; and leukocytosis and cytokine release syndrome in one patient.

Figure 2. Change in Tumor Burden

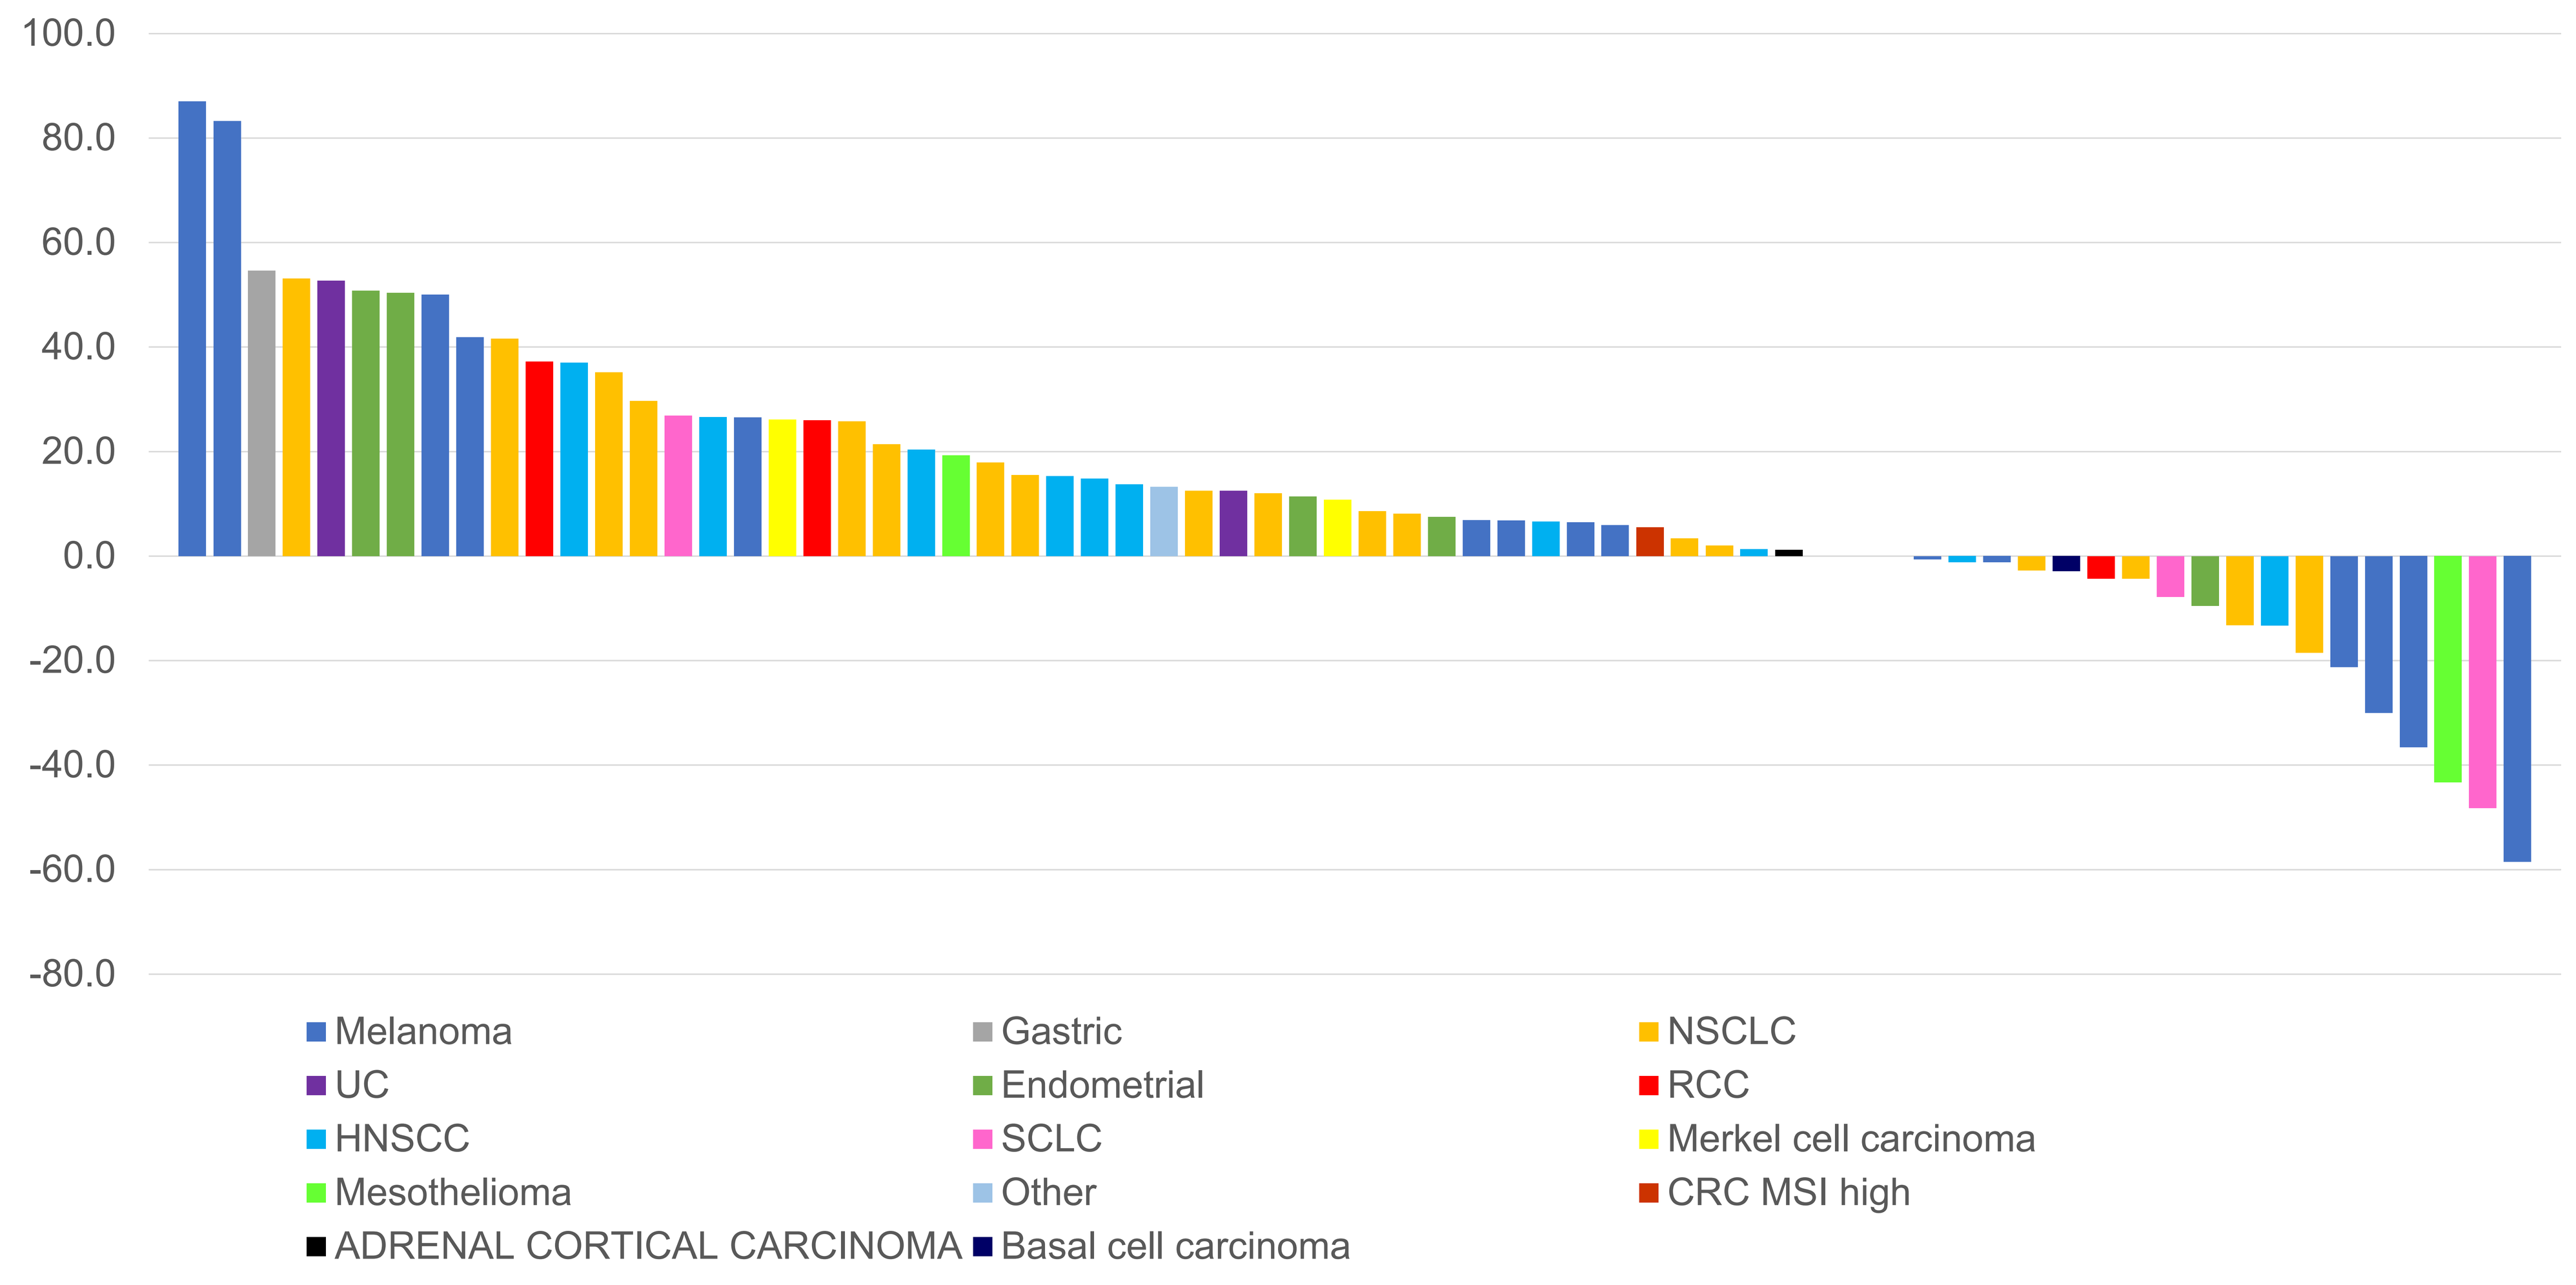
100.0
80.0
60.0
40.0
20.0
0.0
-20.0
-40.0
-60.0
-80.0
Melanoma
UC
HNSCC
Mesothelioma
ADRENAL CORTICAL CARCINOMA
Gastric
Endometrial
SCLC
Other
Basal cell carcinoma
NSCLC
RCC
Merkel cell carcinoma
CRC MSI high

Figure 2. In the Dose Expansion portion of the study where two doses were compared (0.3 mg/kg and 0.6 mg/kg), 5 responses were observed. A Complete Response (CR) was confirmed by PET scan in 1 of 3 patients treated with small-cell lung cancer. Four additional PRs were also observed: 3 of 11 (27.3%) patients with melanoma (2 confirmed, one unconfirmed) and 1 of 4 (25%) patients with mesothelioma (PR confirmed).

Figure 3. CR in a patient with small cell lung cancer

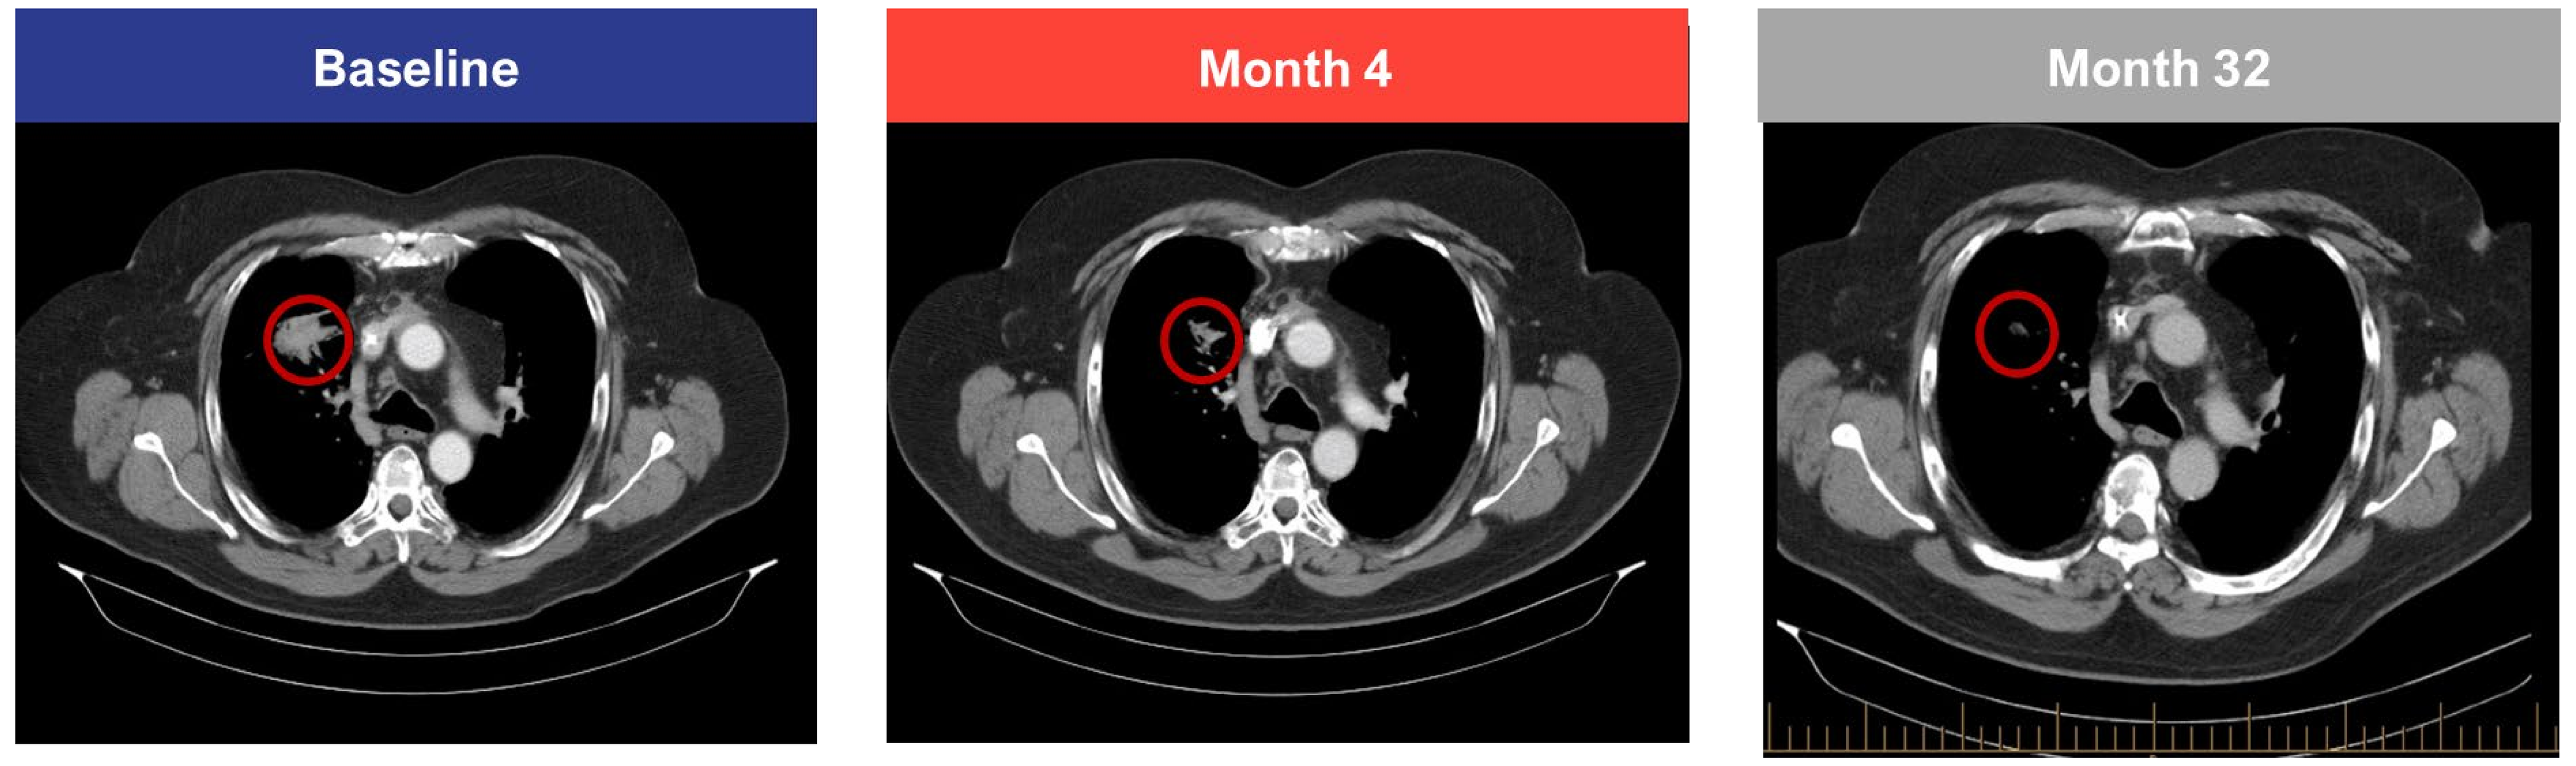
Baseline
Month 4
Month 32

Figure 3. This patient is a 66 y.o man with metastatic small cell lung cancer. The patient received first-line carboplatin, etoposide, and atezolizumab followed by nivolumab second line. At study entry, the patient had innumerable bone metastases. On CTX-471, he had a confirmed PR at Month 6 that was durable for approximately 3 years prior to converting to a PET-negative CR. All bone metastases and the regressed right upper lobe mass were tracer negative. The patient came off study after 3.5 years and remains with no evidence of disease (NED).

Figure 4. CTX-471 Pharmacokinetics

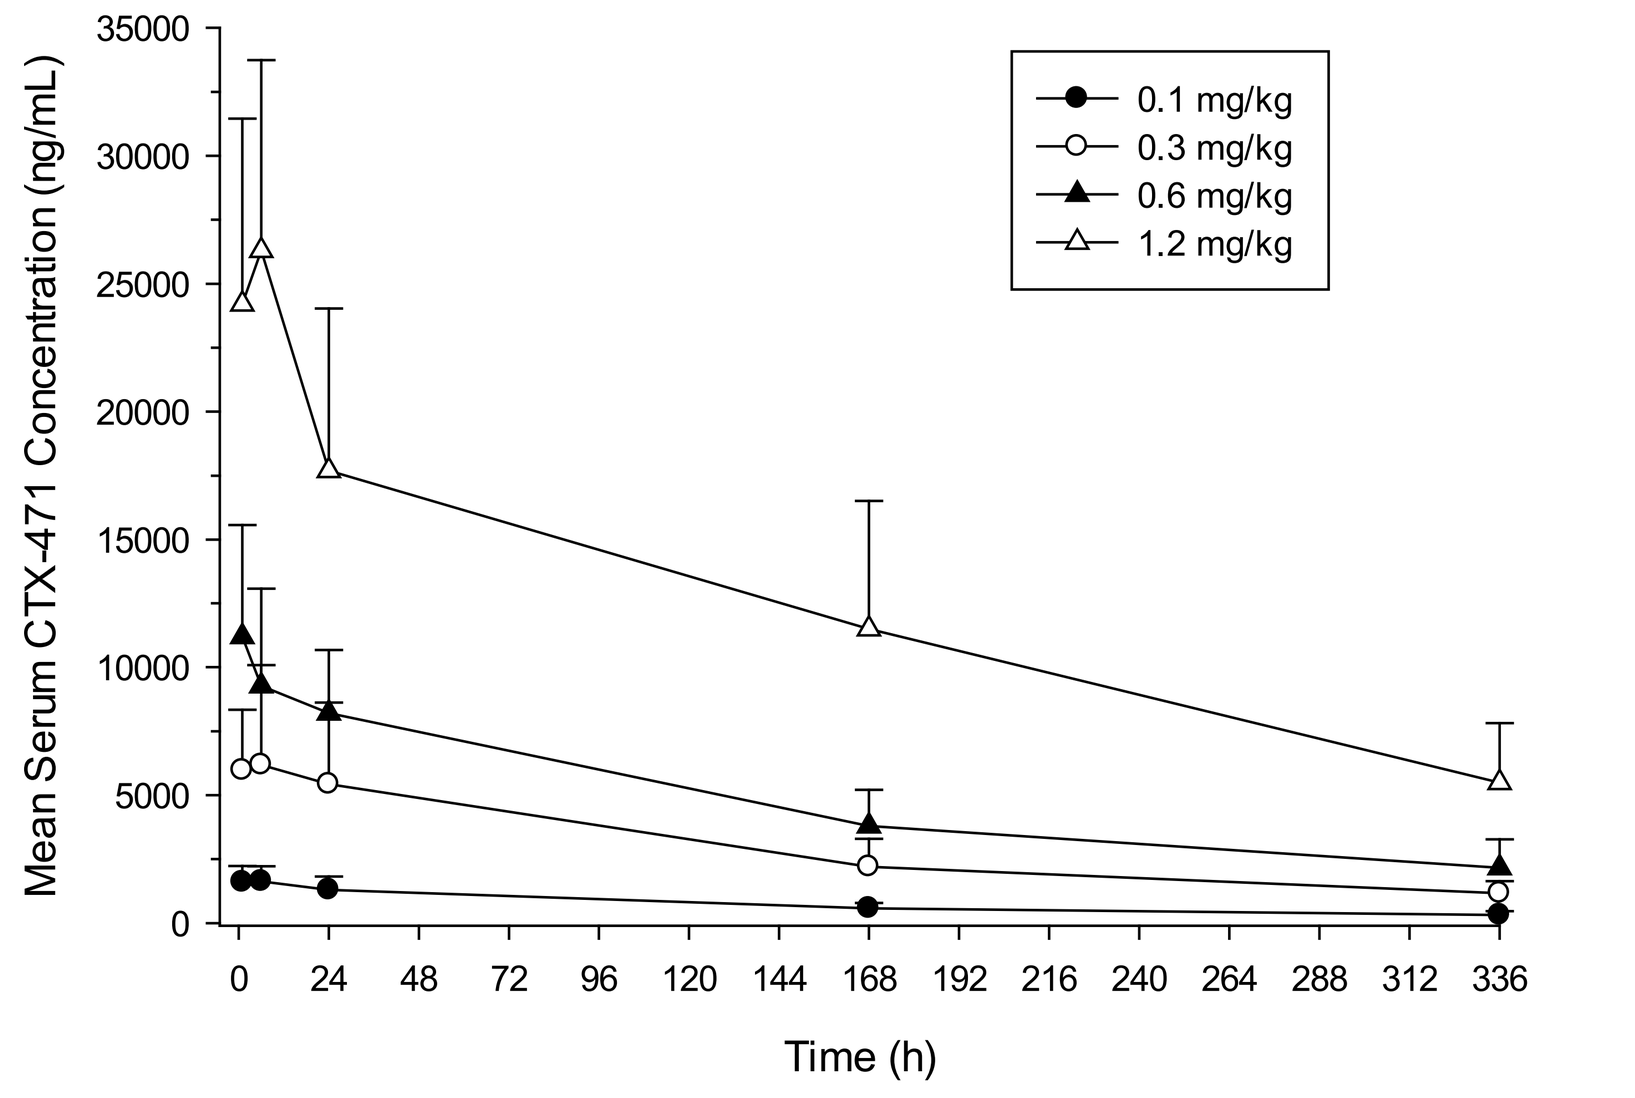
Mean Serum CTX-471 Concentration (ng/mL)
35000
30000
25000
20000
15000
10000
5000
0
0.1 mg/kg
0.3 mg/kg
0.6 mg/kg
1.2 mg/kg
0 24 48 72 96 120 144 168 192 216 240 264 288 312 336
Time (h)

Dose (mg / kg)	Half-life (h)	Cmax (ng/ml)
0.1	147	1,630
0.3	166	7,030
0.6	150	11,900
1.2	196	26,300

Figure 4. CTX-471 pharmacokinetics were measured at first dose on W1D1. Four doses were administered yielding an average half life of approximately 7 days and a dose-dependent Cmax.

Conclusions

In this first-in-human Phase 1 Dose-Escalation and Dose-Expansion Study, CTX-471 monotherapy, observations included:

- ➢ A well-tolerated safety profile
- ➢ A low incidence (6.3%) of LFT increases
- ➢ Expected pharmacokinetics.
- ➢ Very promising evidence of monotherapy anti-tumor activity in refractory patients who had progressed on approved PD-1 or PD-L1 inhibitors.
- ➢ A CR in a patient with small cell lung cancer and 3/11 PRs (27%) in patients with melanoma in the dose expansion phase.

Acknowledgements

- Patients
- Physicians and Study Site Staffs
- Compass Therapeutics Team
- IQVIA Team

Presented at the American Society of Clinical Oncology Annual Meeting 2024, May 31 – June 4, Chicago, IL, USA & Online, Abstract #2535